Primary Diagnosis of Breast Biopsies supported by AI versus Microscope: Multi-Site Clinical Reader Study

Anne Vincent-Salomon[1] | Alona Nudelman[2] | Joanna Cyrta[1] | Marina Maklakovski[3] | Anat Albrecht Shach[4]
Geraldine Sebag[5] | Giuseppe Mallel[5] | Ira Krasnitsky[5] | Tali Feinberg[2] | Manuela Vecsler[5] | Judith Sandbank[2,5]

[1]Diagnostic and Theranostic Medicine Division, Department of Pathology, Institut Curie, PSL University, Paris, France; [2]Institute of Pathology, Maccabi Healthcare Services, Rehovot, Israel; [3]Department of Pathology, Assuta Ashdod Medical Center, Ashdod, Israel; [4]Department of Pathology, Shamir Medical Center, Beer Yaakov, Israel; [5]Ibex Medical Analytics, Tel Aviv, Israel

Contact | anne.salomon@curie.fr

BACKGROUND

- Breast cancer survival rates have been repeatedly correlated with early detection of the disease in its localized stage[1].
- Pathological diagnosis of breast cancer is complicated due to a variety of morphologic features including tumor growth pattern and nuclear cytologic features, yet its accuracy is of utmost importance as the pathologic assessment dictates the clinical management of the patient and provides prognostic information.
- Artificial intelligence (AI)-assisted analysis of digitized images of pathology biopsy slides has demonstrated high accuracy in the detection of cancer, leading in turn to better patient outcomes[2,3].

OBJECTIVE

- The objective of the study was to demonstrate the non-inferiority of the Galen Breast solution in a primary diagnosis workflow to the standard of care workflow (SOC) in detecting breast cancer, including invasive and in-situ carcinoma in a population of adult female subjects who have undergone a breast biopsy.

METHODS

Galen Breast:

- The solution was developed based on fully-supervised, multilayered convolutional neural networks (CNNs) designed to classify and analyze whole slide images (WSI).
- Trained on more than 2 million labeled images from samples from different pathology departments, annotated manually by more than 20 senior pathologists.
- Identifies more than 50 different classes of pathologic and benign breast features.

Study design

- A two-arm clinical reader study included tissue samples retrieved from two sites, processed using different staining protocols and digital scanners.
- Eight pathologists blindly reviewed biopsies in both AI-assisted and SOC (review under microscope) arms.
- Diagnoses were compared to the ground truth (GT) that was established by a consensus of two breast pathologists.
- Major discrepancy rate per case between each arm and the GT was examined, major discrepancy was defined as a difference in diagnosis associated with a difference in patient management and included: Invasive vs. Benign, Invasive vs. DCIS/ADH, DCIS/ADH vs. Benign, IDC vs. ILC vs. other subtypes.

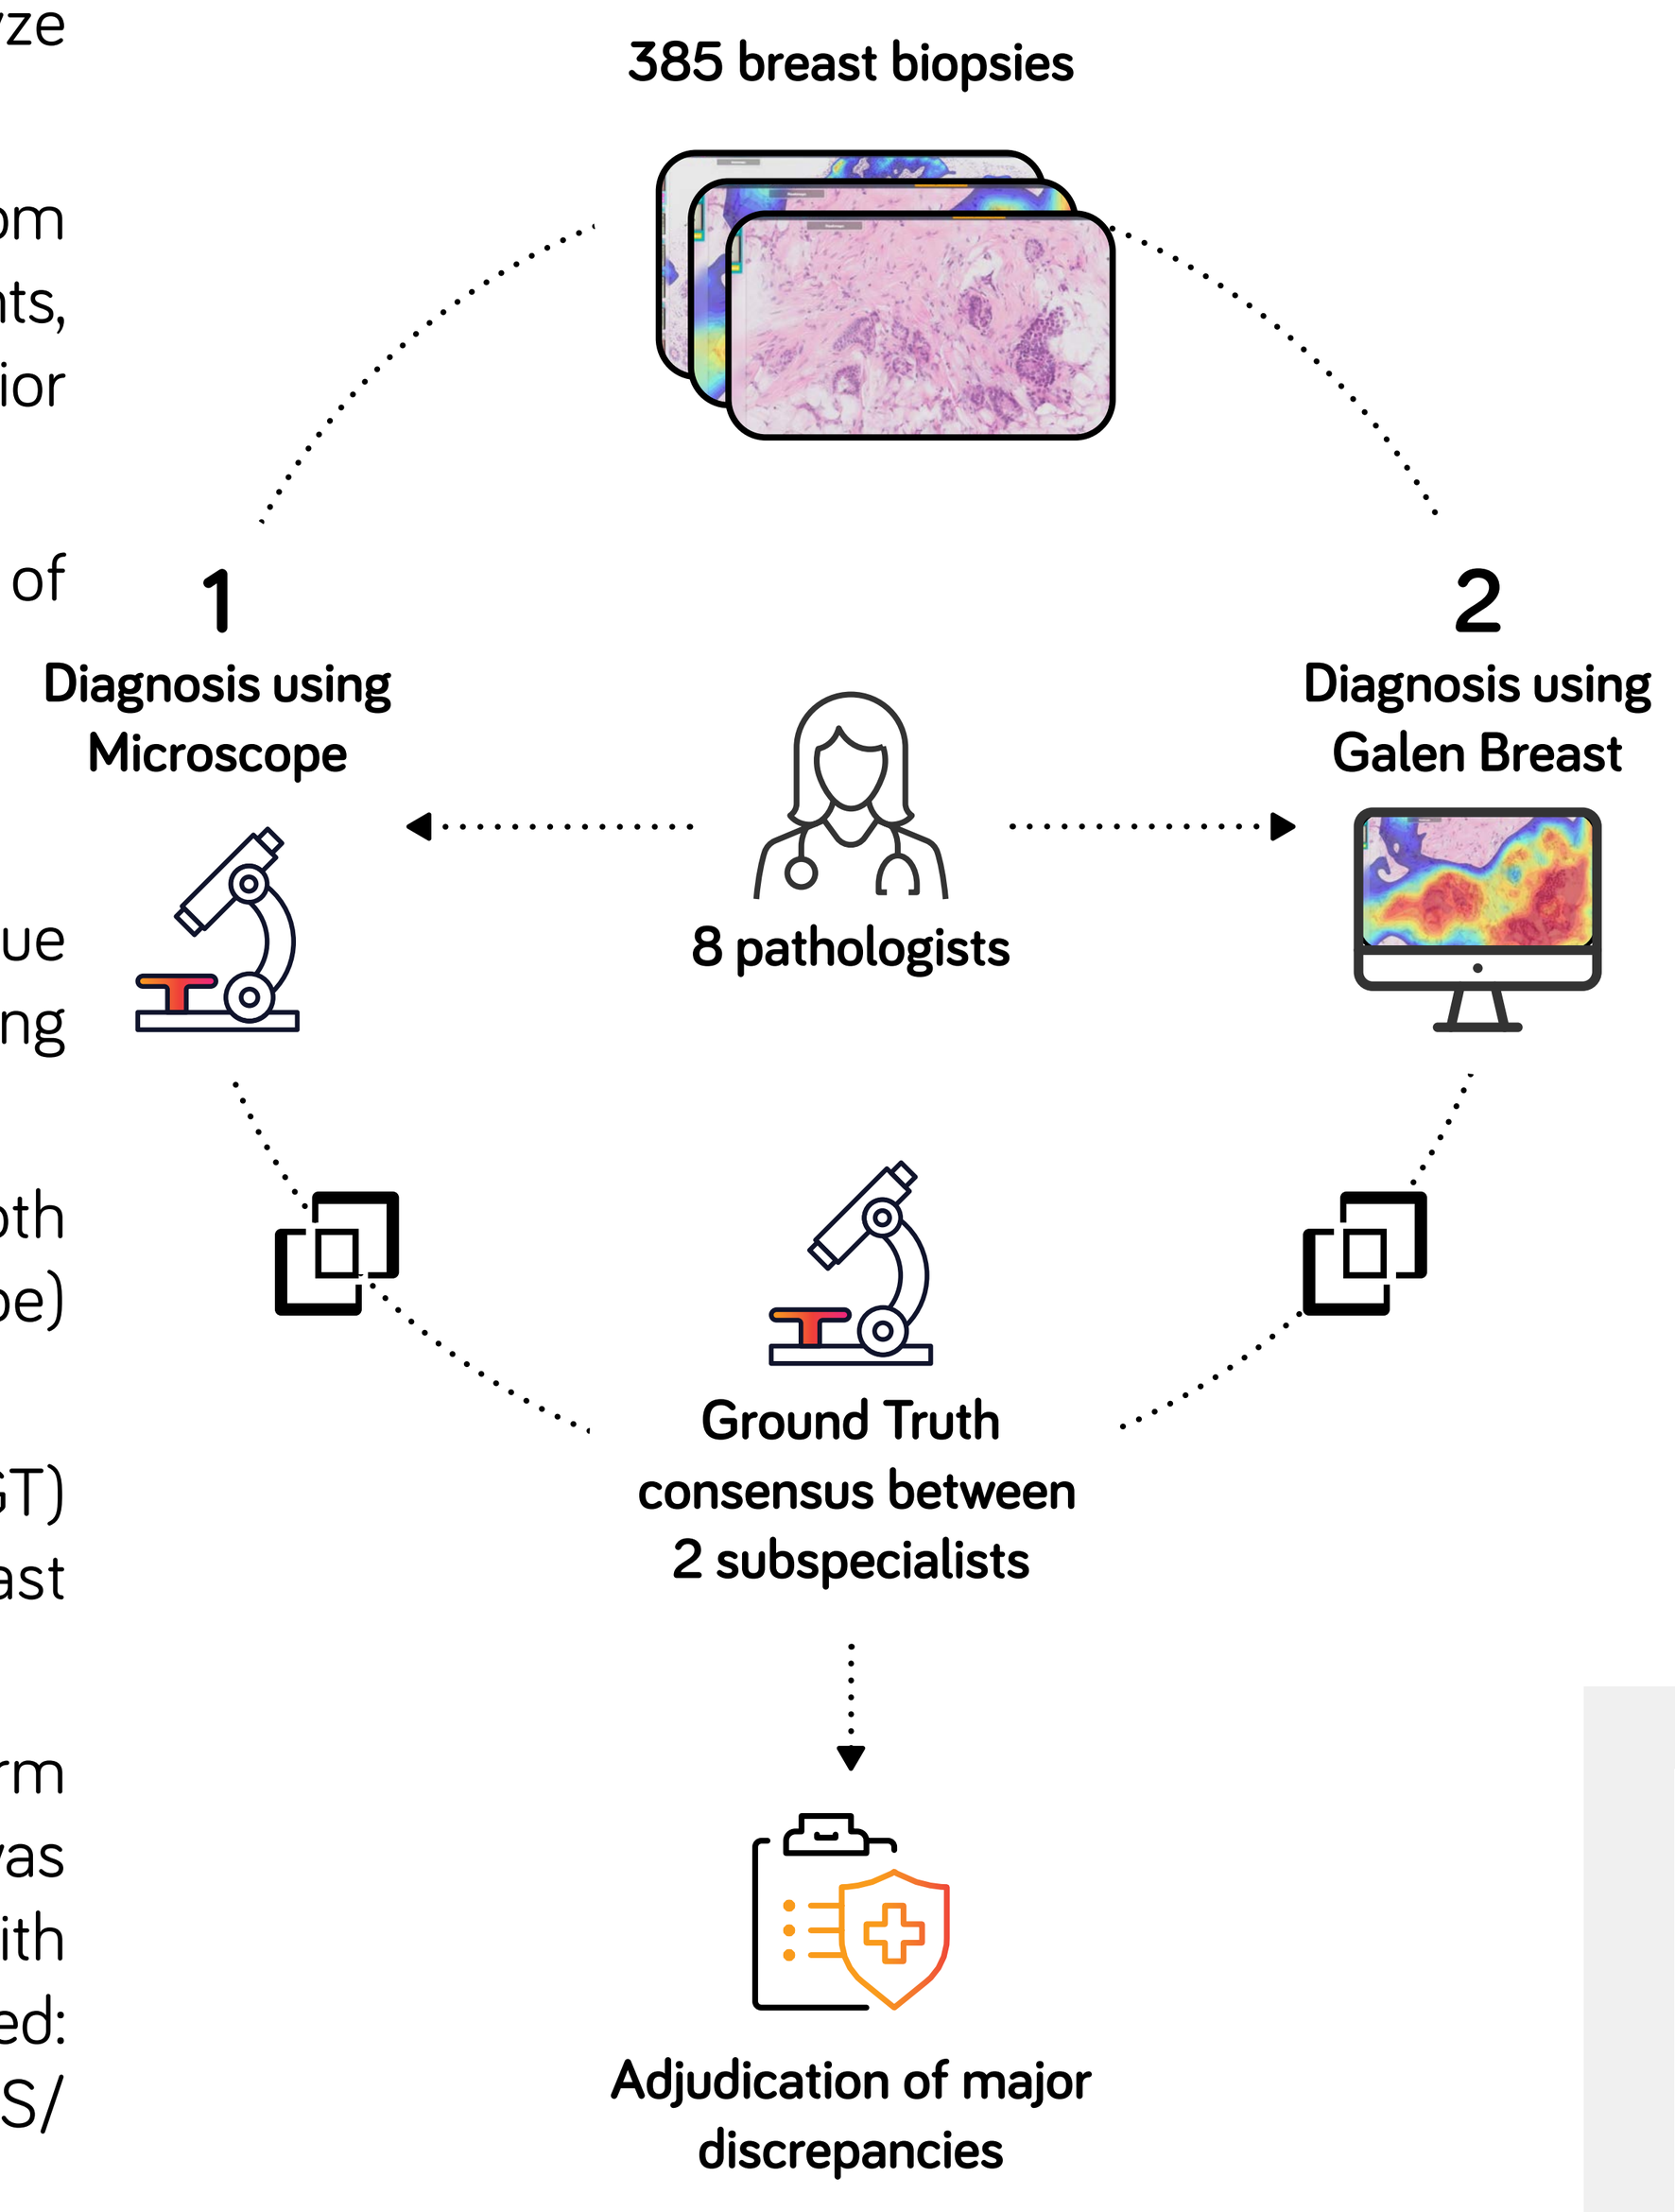
385 breast biopsies
1
Diagnosis using Microscope
2
Diagnosis using Galen Breast
8 pathologists
Ground Truth consensus between 2 subspecialists
Adjudication of major discrepancies

Figure 1 - Study design

Cohort characteristics

	Site 1	Site 2	total
Number of Cases	234	151	385
Mean age at Diagnosis	51.90	53.79	52.65
Number of HES/ H&E slides	442	330	772
Vacuum-assisted biopsy	92 (39.32%)	63 (41.72%)	155 (40.26%)
Core Needle Biopsy	142 (60.68%)	88 (58.28%)	230 (59.74%)

Table 1 – Cohort characteristics

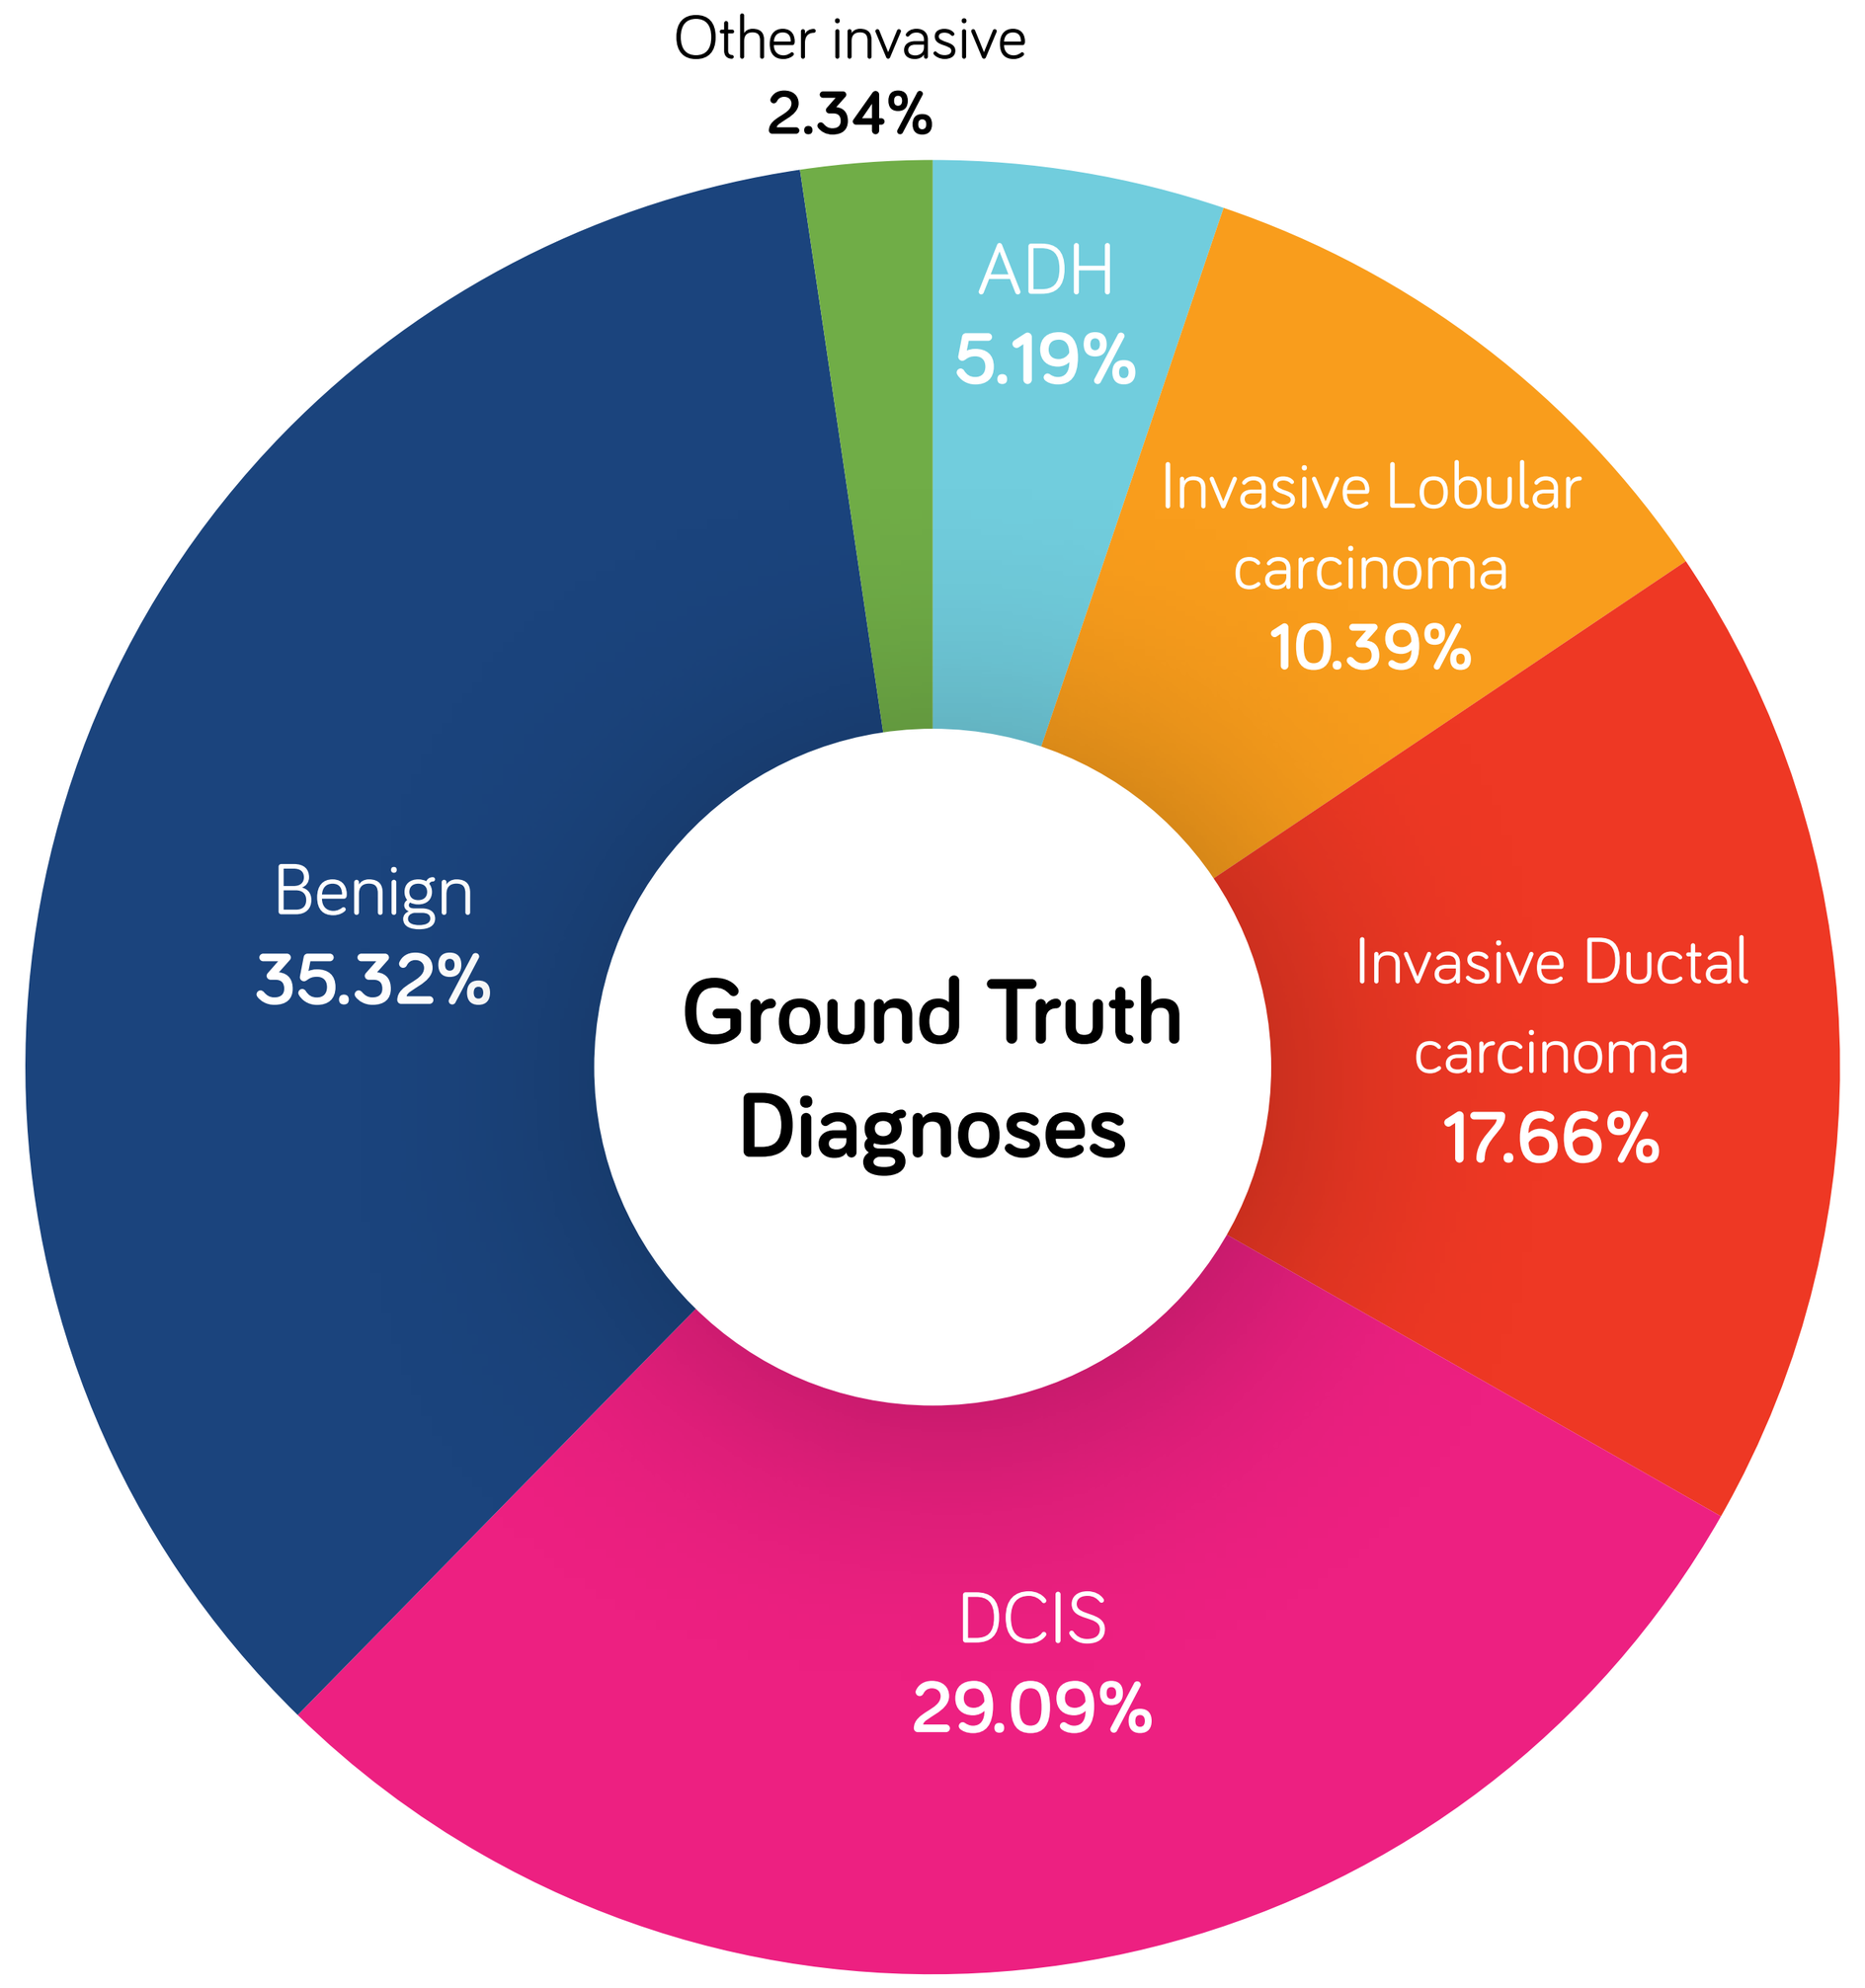
Other invasive
2.34%
ADH
5.19%
Invasive Lobular carcinoma
10.39%
Invasive Ductal carcinoma
17.66%
DCIS
29.09%
Benign
35.32%
Ground Truth Diagnoses

Figure 2 - Distribution of ground truth diagnoses

- Pathologists reported on a total of 772 H&E/HES slides from 385 breast biopsies. Of these cases, 117 were of invasive breast cancer, 132 DCIS/ADH, 136 benign.

RESULTS

Galen Breast solution demonstrated high accuracy in the detection of breast cancer, aiding pathologists in comprehensive review of breast biopsies.

- The major discrepancy rates (across all diagnoses) of the SOC arm and of the AI assisted arm vs. GT were 4.42% and 3.12%, respectively, demonstrating a 29.4% reduction in major discrepancy from GT when assisted by the AI.
- AI- assisted pathologists demonstrated high accuracy in detection of breast pathologies with sensitivity and specificity of 95.98% and 97.79% respectively for the detection of cancer, 100% and 100% respectively for the detection of invasive carcinoma and 92.42% and 97.79% in detection of DCIS/ADH.
- Galen Breast demonstrated excellent performance in detecting DCIS, DCIS grading (high grade vs. low grade/ADH), and IDC vs. ILC as depicted in the ROC curves with AUC of 0.963, 0.921 and 0.982, respectively.

Invasive Carcinoma Detection Assisted by the Galen Breast AI vs. SOC

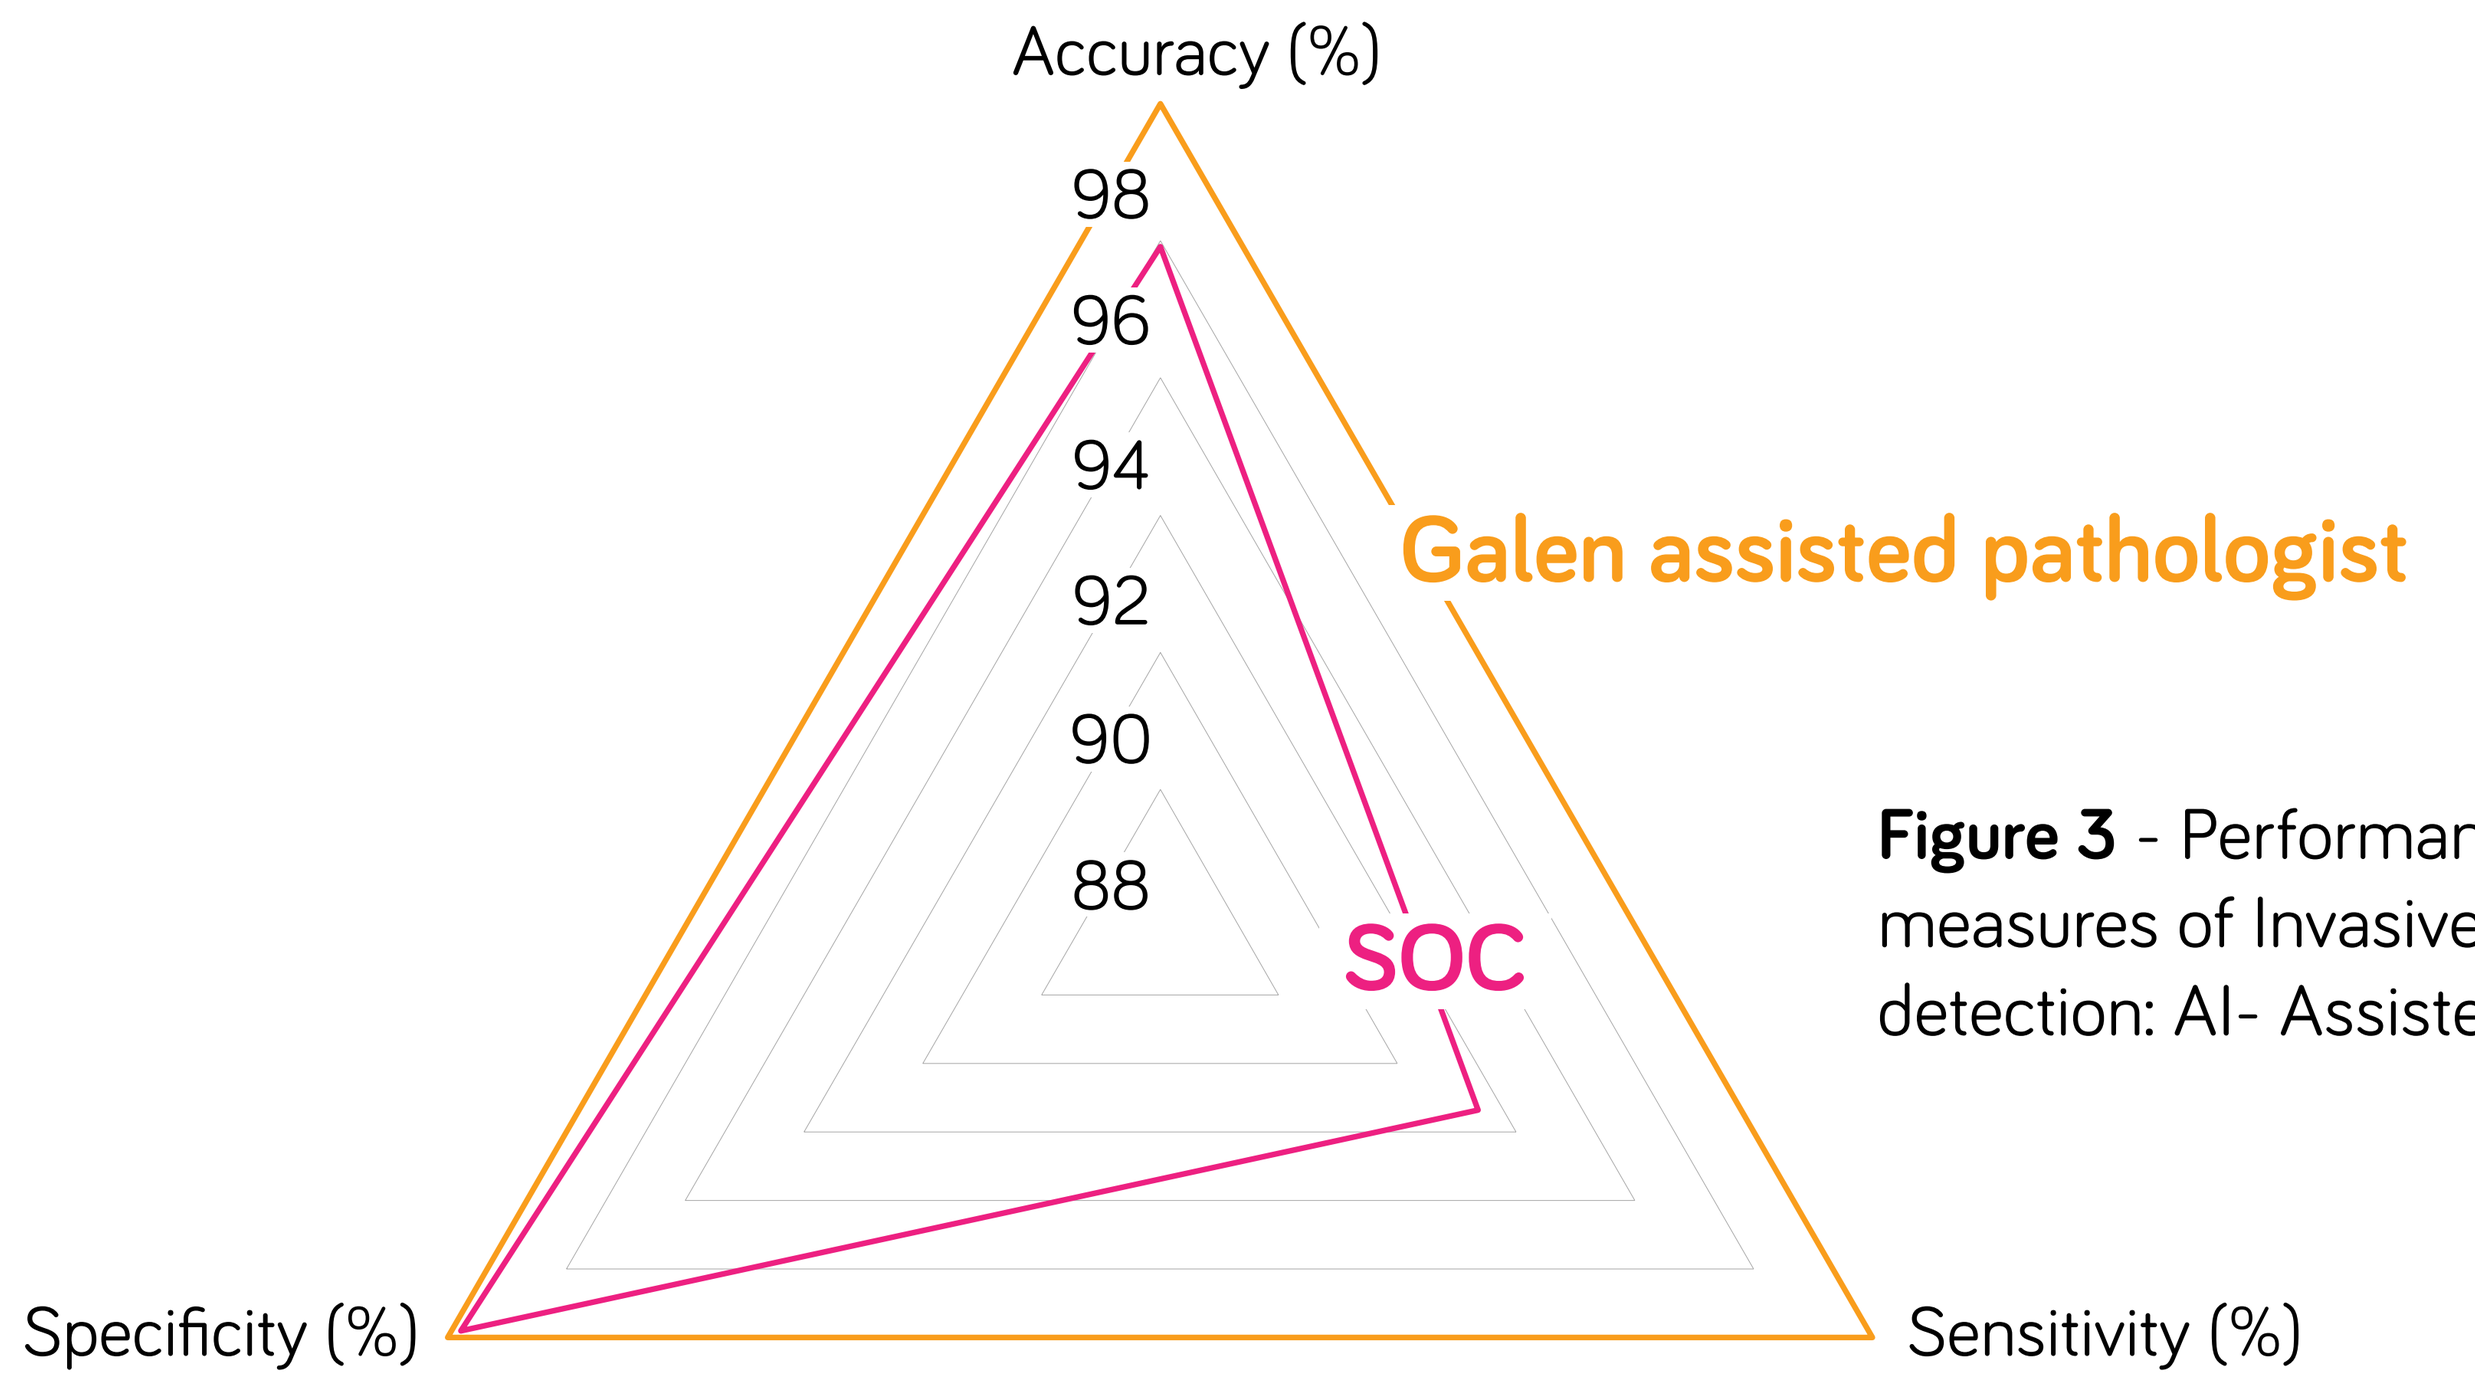
Accuracy (%)
98
96
94
92
90
88
Galen assisted pathologist
SOC
Specificity (%)
Sensitivity (%)

Figure 3 - Performance measures of Invasive carcinoma detection: AI- Assisted vs. SOC

ROC Curves for DCIS detection, grading and IDC vs. ILC

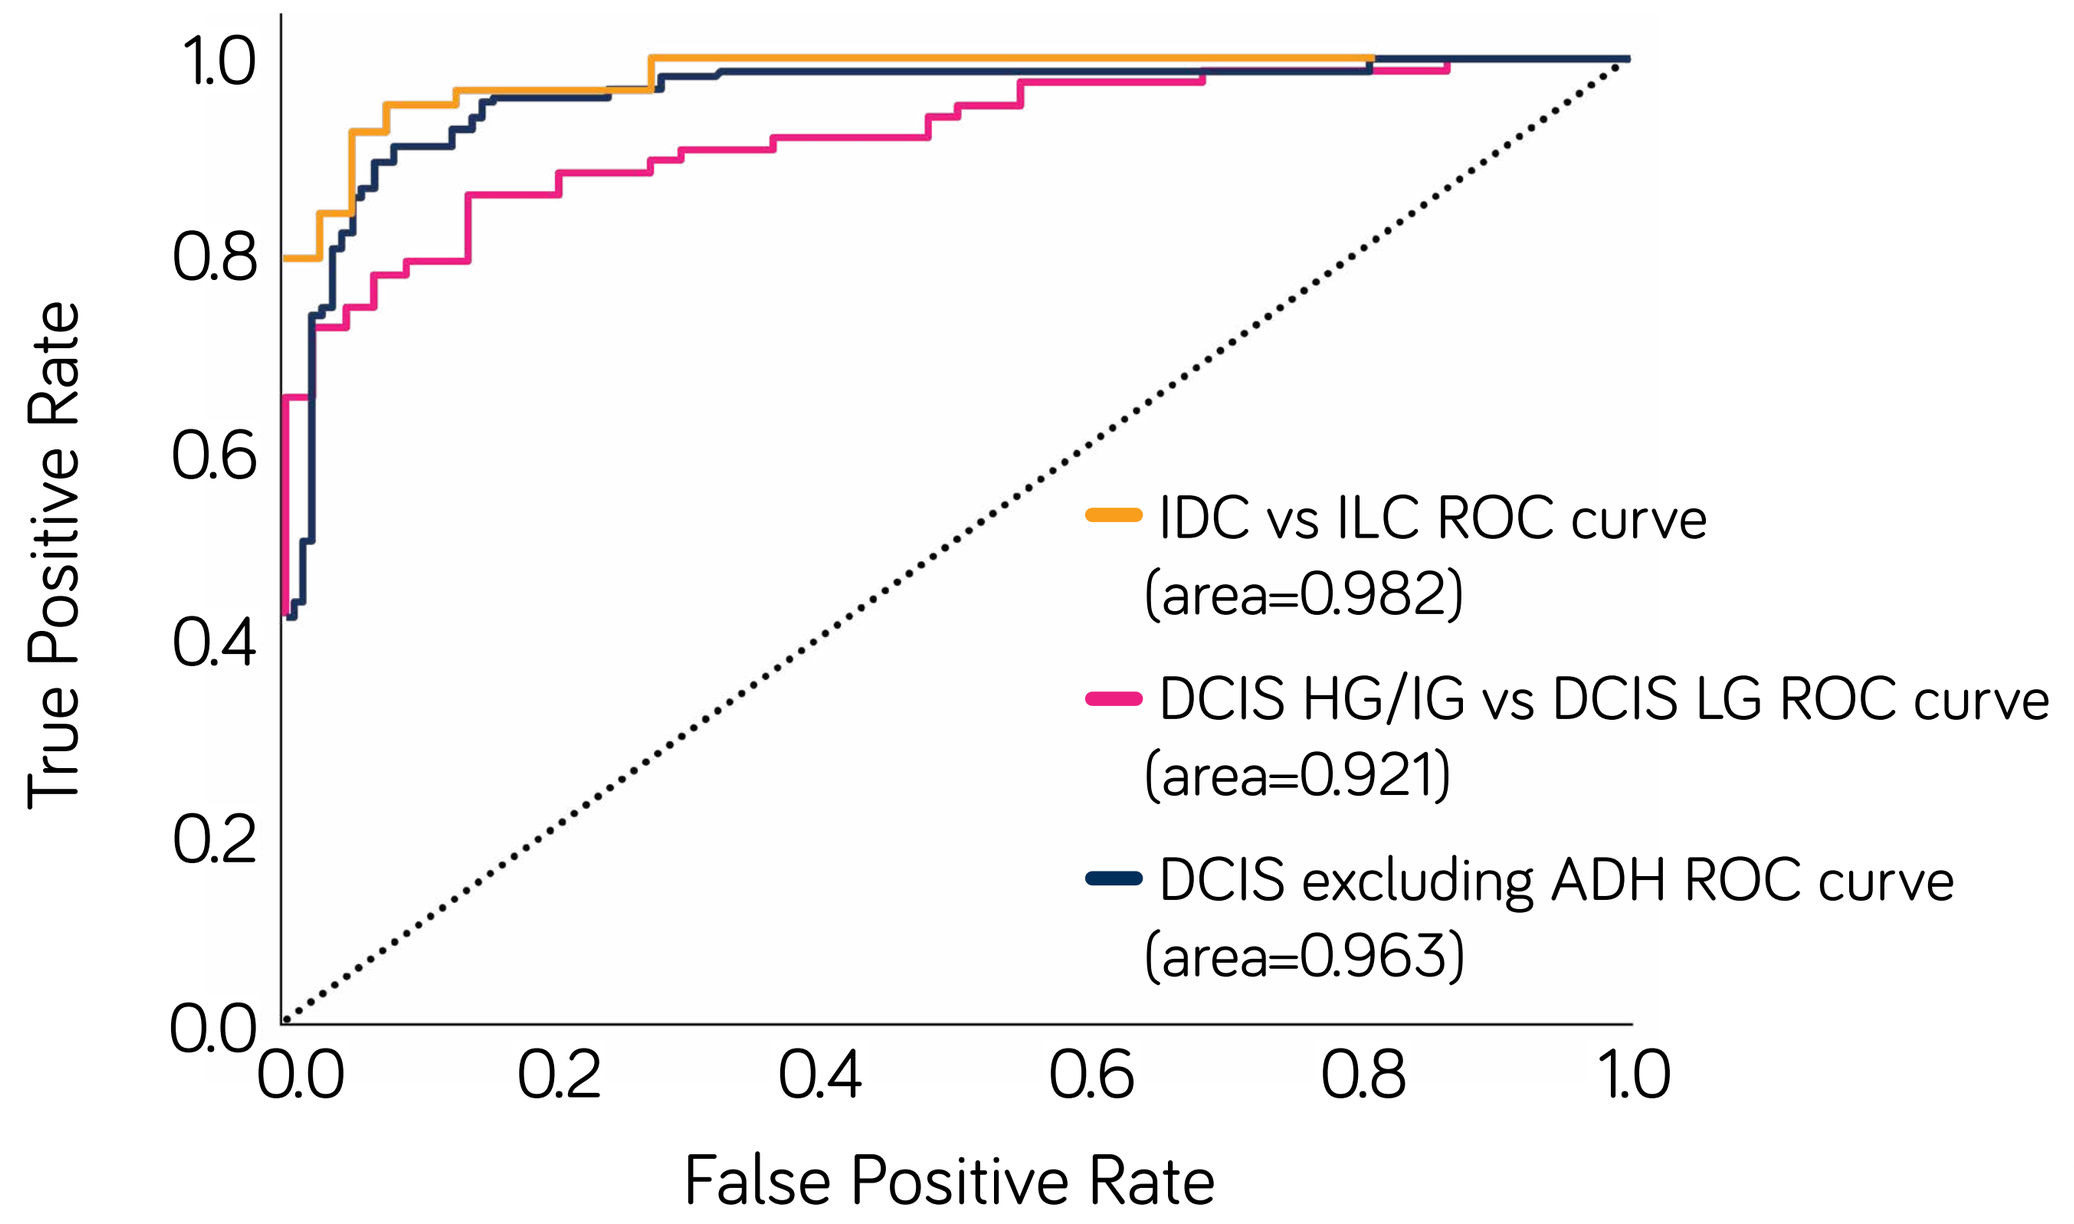
True Positive Rate
False Positive Rate
1.0
0.8
0.6
0.4
0.2
0.0
IDC vs ILC ROC curve (area=0.982)
DCIS HG/IG vs DCIS LG ROC curve (area=0.921)
DCIS excluding ADH ROC curve (area=0.963)

Figure 4 - Performance measures of DCIS detection and grading, IDC Vs. ILC

Pathologists' performance using Galen Breast or SOC

	Major discrepancies N (%)	95% CI
Galen Breast	12 (3.12%)	1.38 - 4.85%
Standard Of Care	17 (4.42%)	2.36 - 6.47 %
Difference	**-29%**	-

Table 2 - Discrepancy rate comparison of the study arms

AI Assisted pathologist identifies IDC that was missed on the microscope

Figure 5 | A | IDC Tubular, H&E (X8) **B |** IDC Tubular with Invasive Heatmap (X8) **C |** IDC Tubular, IHC Myosin (X8)

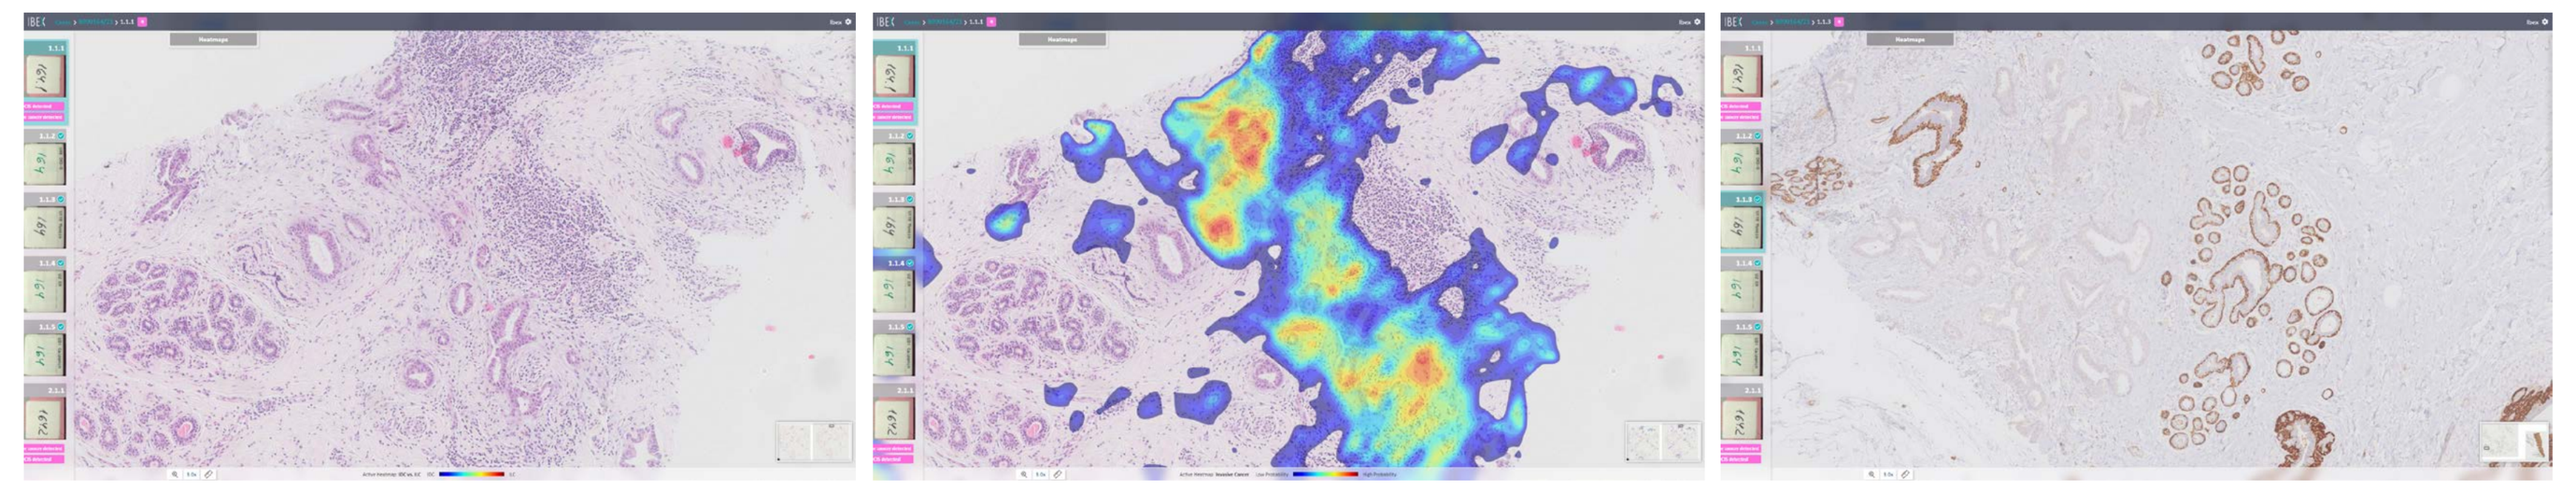

AI Assisted pathologist identifies small focus of IDC missed on the microscope

Figure 6 | A | IDC, Invasive Cancer Heatmap (X0.8) **B |** Zoom on Invasive Focus, H&E (X18) **C |** IDC, Zoom on Invasive Focus, Invasive Heatmap (X18)

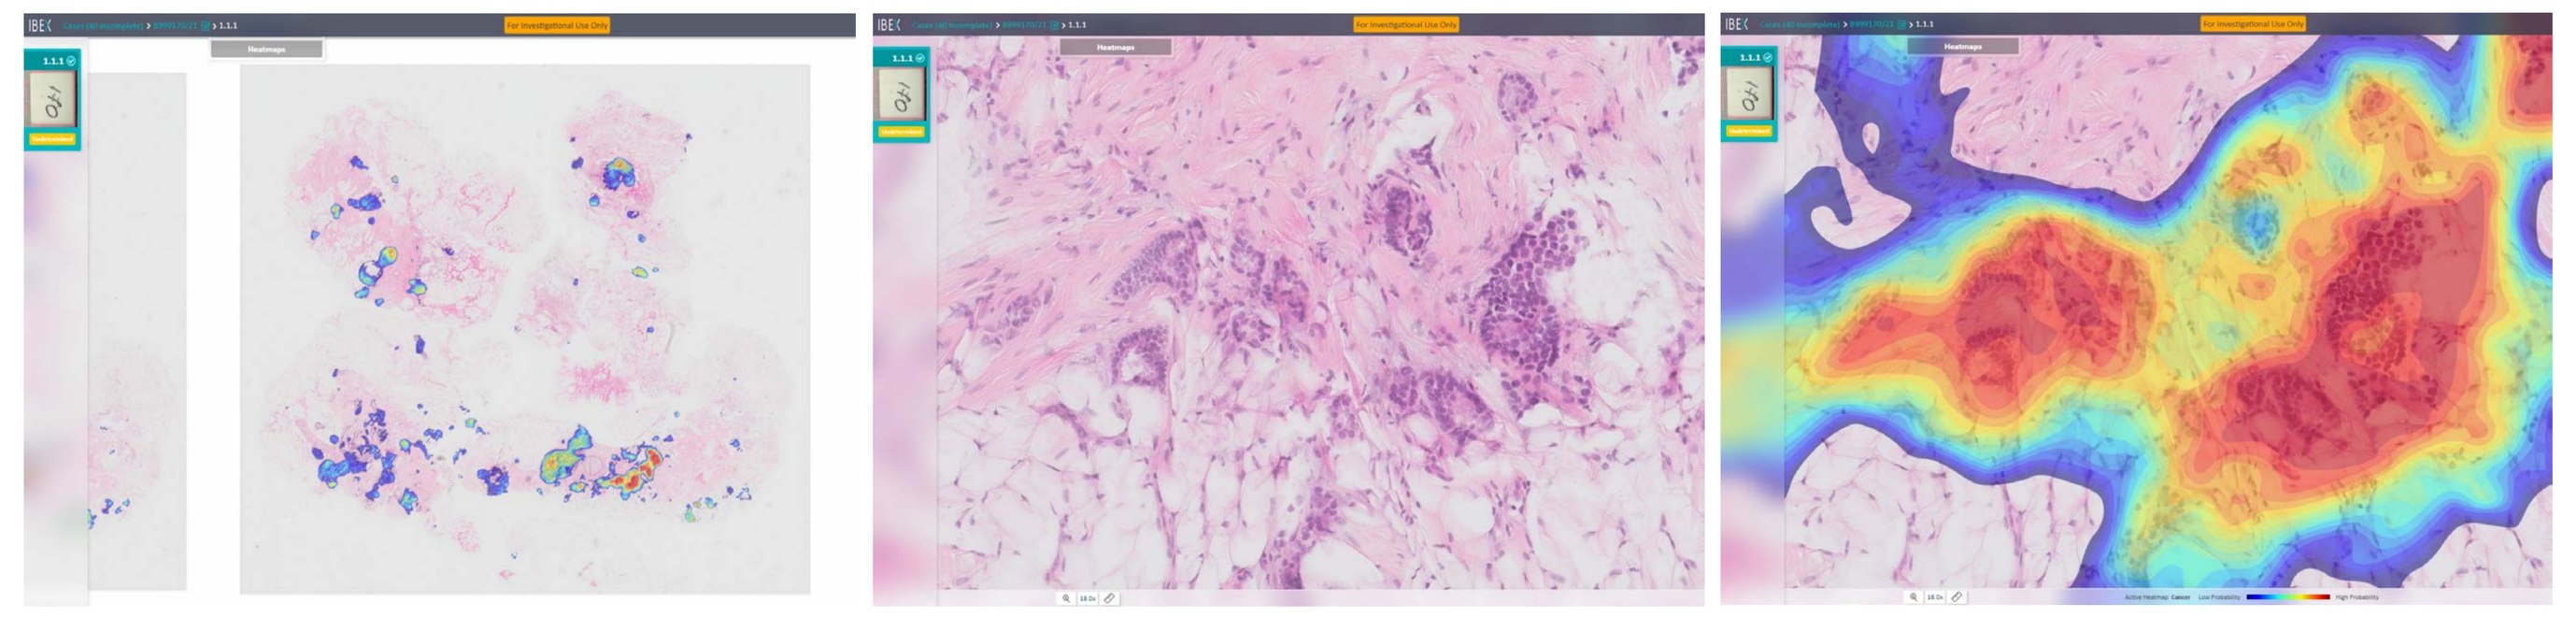

High performance of Galen Breast in DCIS detection

Figure 7 | A | DCIS Heatmap (X1) **B |** DCIS, H&E (X8) **C |** DCIS Heatmap (X8)

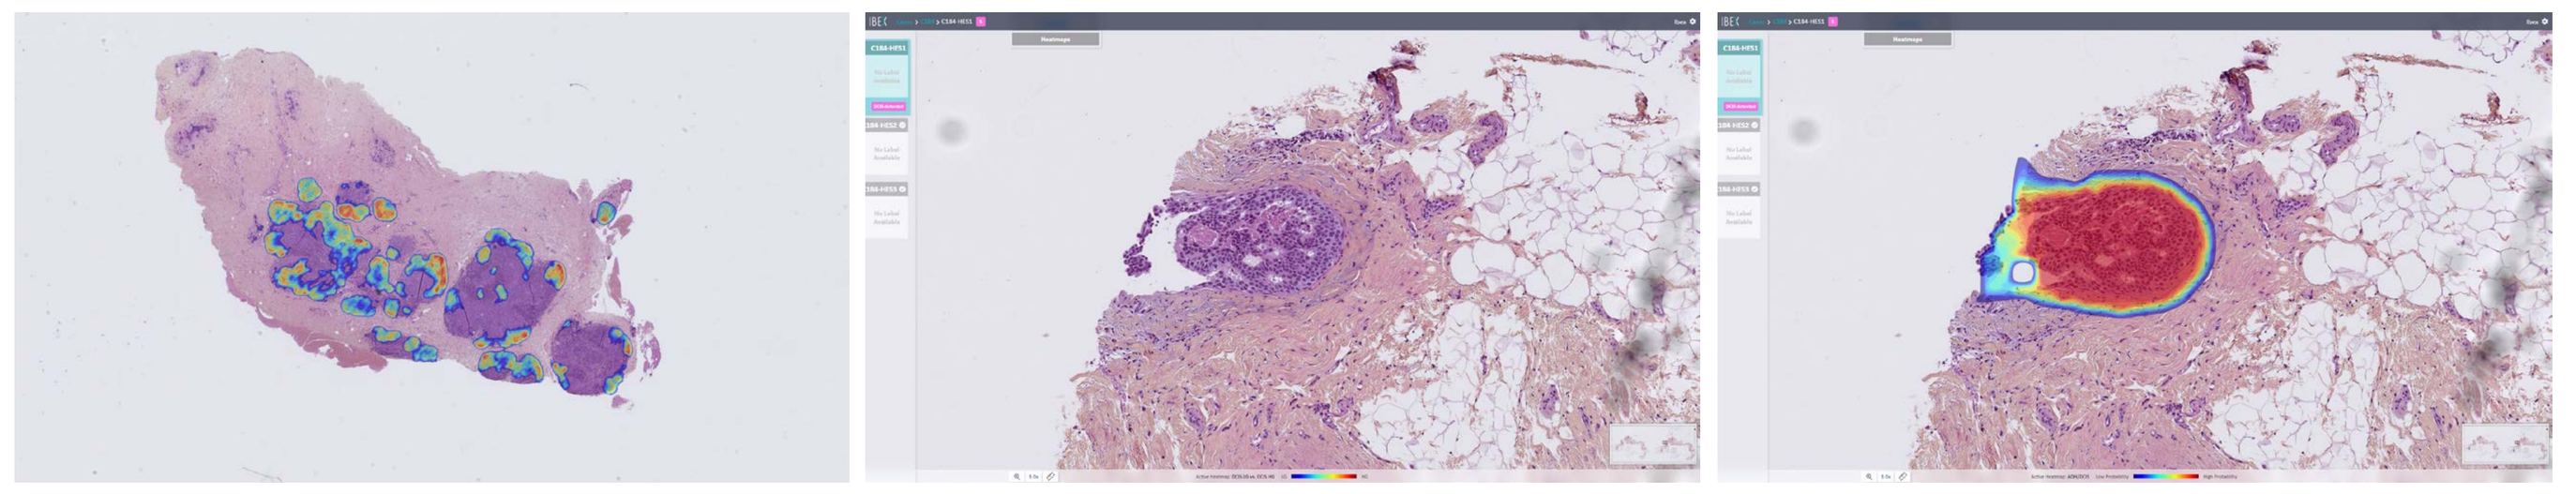

CONCLUSIONS

- Galen Breast improved pathologists' diagnostic quality, resulting in **29% decrease in major discrepancy rate.**
- Detection of invasive carcinoma by AI-assisted pathologist was significantly increased, reflected in reaching **sensitivity of 100%.**
- The solution demonstrated high accuracy rate across different geographies, different scanners and different staining protocols
- This study demonstrated AI's ability to support pathologists with an efficient workflow and multiple diagnostic insights that would allow for increased diagnostic capacity and improved patient care

References : [1]https://www.cancer.net/cancer-types/breast-cancer/statistics, [2]Vincent-Salomon et al., Modern Pathology 2022, 35(S2):203-204 3 Sandbank et al., Modern Pathology 2022, 35(S2):1094