A fully automatic artificial intelligence system for accurate and reproducible HER2 IHC scoring in breast cancer

Yuval Globerson[1] | Lilach Bien[1] | Jonathan Harel[1] | Giuseppe Mallel[1] | Geraldine Sebag[1] | Michel Vandenberghe[2] | Craig Barker[2] | Tsuyoshi Matsuo[2] | Charo Garrido[3] | Maya Grinwald[1] | Manuela Vecsler[1] |Judith Sandbank[1] | Chaim Linhart[1]

[1]Ibex Medical Analytics, Tel Aviv, Israel; [2]AstraZeneca, Cambridge, UK; [3]Daiichi Sankyo, Basking Ridge, NJ, USA. Contact | chaim.linhart@ibex-ai.com

Acknowledgments | This study is sponsored by Ibex Medical Analytics and AstraZeneca Pharmaceuticals in collaboration with Daiichi Sankyo, Inc.
Disclosures | Yuval Globerson, Lilach Bien, Jonathan Harel, Giuseppe Mallel, Geraldine Sebag, Maya Grinwald, Manuela Vecsler, Judith Sandbank, Chaim Linhart are employees of Ibex Medical Analytics and have an ownership interest in stock options. Michel Vandenberghe, Tsuyoshi Matsuo, Craig Barker are employees of AstraZeneca and have an ownership interest in stocks/stock options. Charo Garrido is an employee of Daiichi Sankyo, Inc, and has ownership interest in stocks/stock options.

BACKGROUND

- Breast cancer is the most common malignant disease worldwide and tumor human epidermal growth factor receptor 2 (HER2) expression is one of its key prognostic and treatment influencing factors.
- Visual interpretation of HER2 immunohistochemistry (IHC) expression is subjective, which leads to intra- and inter-pathologist variability[1,2].
- Enhertu (trastuzumab deruxtecan) was recently approved by US FDA for patients with HER2-low metastatic breast cancer (HER2 IHC1+ or IHC2+/ISH-), thus raising the need for accurate and reproducible scoring[3].

OBJECTIVE

To develop and test the performance of an artificial intelligence (AI)-based algorithm for HER2 scoring according to 2018 ASCO/CAP guidelines[4] against rigorous ground truth (GT) established by 3 blinded expert pathologists.

METHODS

Algorithm development A fully automated AI-based solution* for HER2 scoring according to the 2018 ASCO/CAP guidelines was developed. The solution runs three main steps:

Regions model: Deep Learning networks, which classify tissue regions as invasive carcinoma, ductal carcinoma in situ (DCIS) and other morphologies.

- The AI was trained using >750,000 image areas that were extracted from manual and automated annotations in 1,462 slides collected from multiple labs and scanned by 5 different scanners from different manufacturers. Manual annotations were performed by 8 pathologists. Automated annotations were generated by using a previously developed AI model for breast H&E slides classification[5] and projecting its findings from the corresponding H&E slides to the HER2 IHC slides using a registration algorithm.

Cells model: Object Detection networks process only the regions classified as invasive cancer, detect the tumor cells within them and classify their staining pattern (e.g., not stained, moderate incomplete, etc.).

- A total of 350,000 cells were annotated in 230 slides by 8 pathologists.
- Training slides represent all HER2 scores and various types/subtypes of carcinoma.

Score calculation: Detected cells are counted, and the 2018 ASCO/CAP guidelines are applied to derive the slide-level HER2 score.

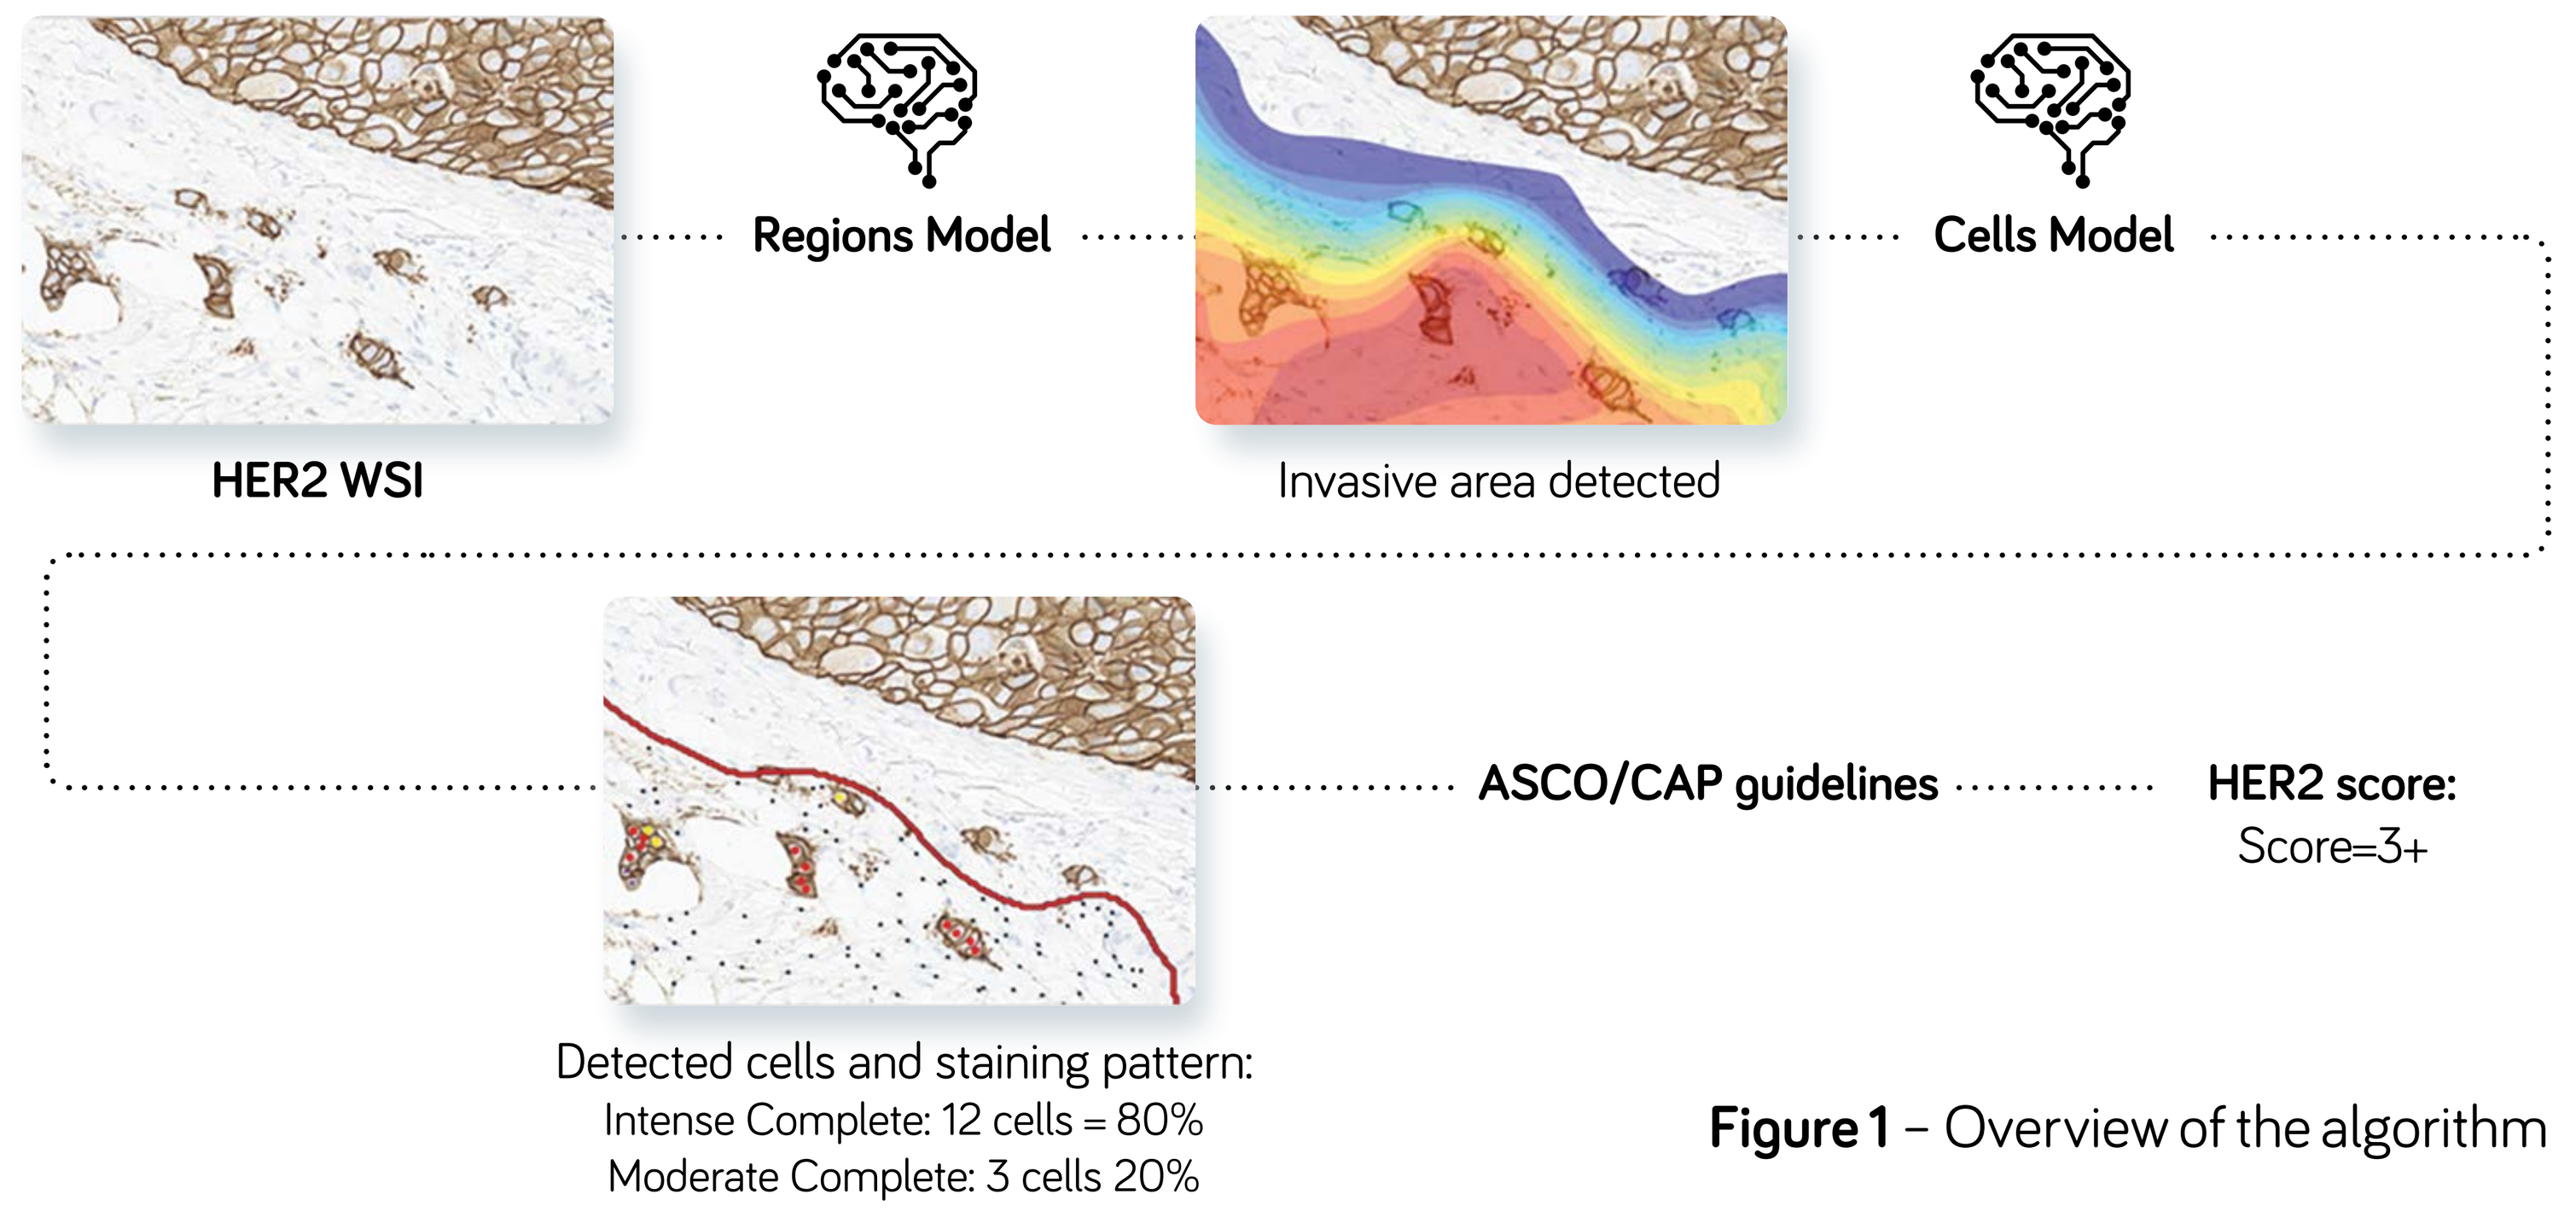
HER2 WSI
Regions Model
Invasive area detected
Cells Model
ASCO/CAP guidelines
HER2 score:
Score=3+
Detected cells and staining pattern:
Intense Complete: 12 cells = 80%
Moderate Complete: 3 cells 20%

Figure 1 – Overview of the algorithm

* The solution is for research use only (RUO)

References: 1. Schettini F, et al. NPJ Breast Cancer. 2021, 7(1):1; 2. Gavrielides et al, Arch Pathol Lab Med, 2011, 135:233–242 ; 3. Modi et al, NEJM, 2022; 387:9; 4. Wolff AC, et al, Arch Pathol Lab Med. 2018, 142(11):1364-1382; 5. Vincent-Salomon et al., Modern Pathology 2022, 35(S2):203-204.

RESULTS

Algorithm Validation Dataset

- Validation dataset included HER2 IHC slides stained using anti-HER2/neu rabbit monoclonal primary antibody (4B5, Ventana) as per manufacturer's instructions.
- The dataset included 452 IHC slides with primary tumor; for 435 slides a GT score was determined.
- For 16 slides with GT the AI indicated that it could not reach a reliable score. For 1 slide the AI didn't detect a sufficient number of tumor cells for reliable scoring. The other 15 slides received borderline scores (e.g. 1+/2+) - in all of these slides, one of the two HER2 scores suggested by the AI matched the GT.

Category			# Cases	% of cases
Type of Specimen	Core-needle biopsies		361	86.2
	Excisions		58	13.8
Type of Cancer	Invasive Ductal Carcinoma (IDC)		343	81.9
	Invasive Lobular Carcinoma (ILC)		30	7.2
	IDC + ILC		16	3.8
	Other Invasive		30	7.2
Lab and Scanner	Lab A	Leica GT450 Leica CS2	142	33.9
	Lab B	Leica GT450 Leica Scanscope XT	149	35.6
	Lab C	Leica GT450	128	30.5
Total			419	

Table 1 – Validation dataset characteristics

Ground Truth was established by the majority scores of a panel of three pathologists who scored according to the 2018 ASCO/CAP guidelines[4] without additional clinical considerations.

Manual HER2 scoring - agreement among GT pathologists

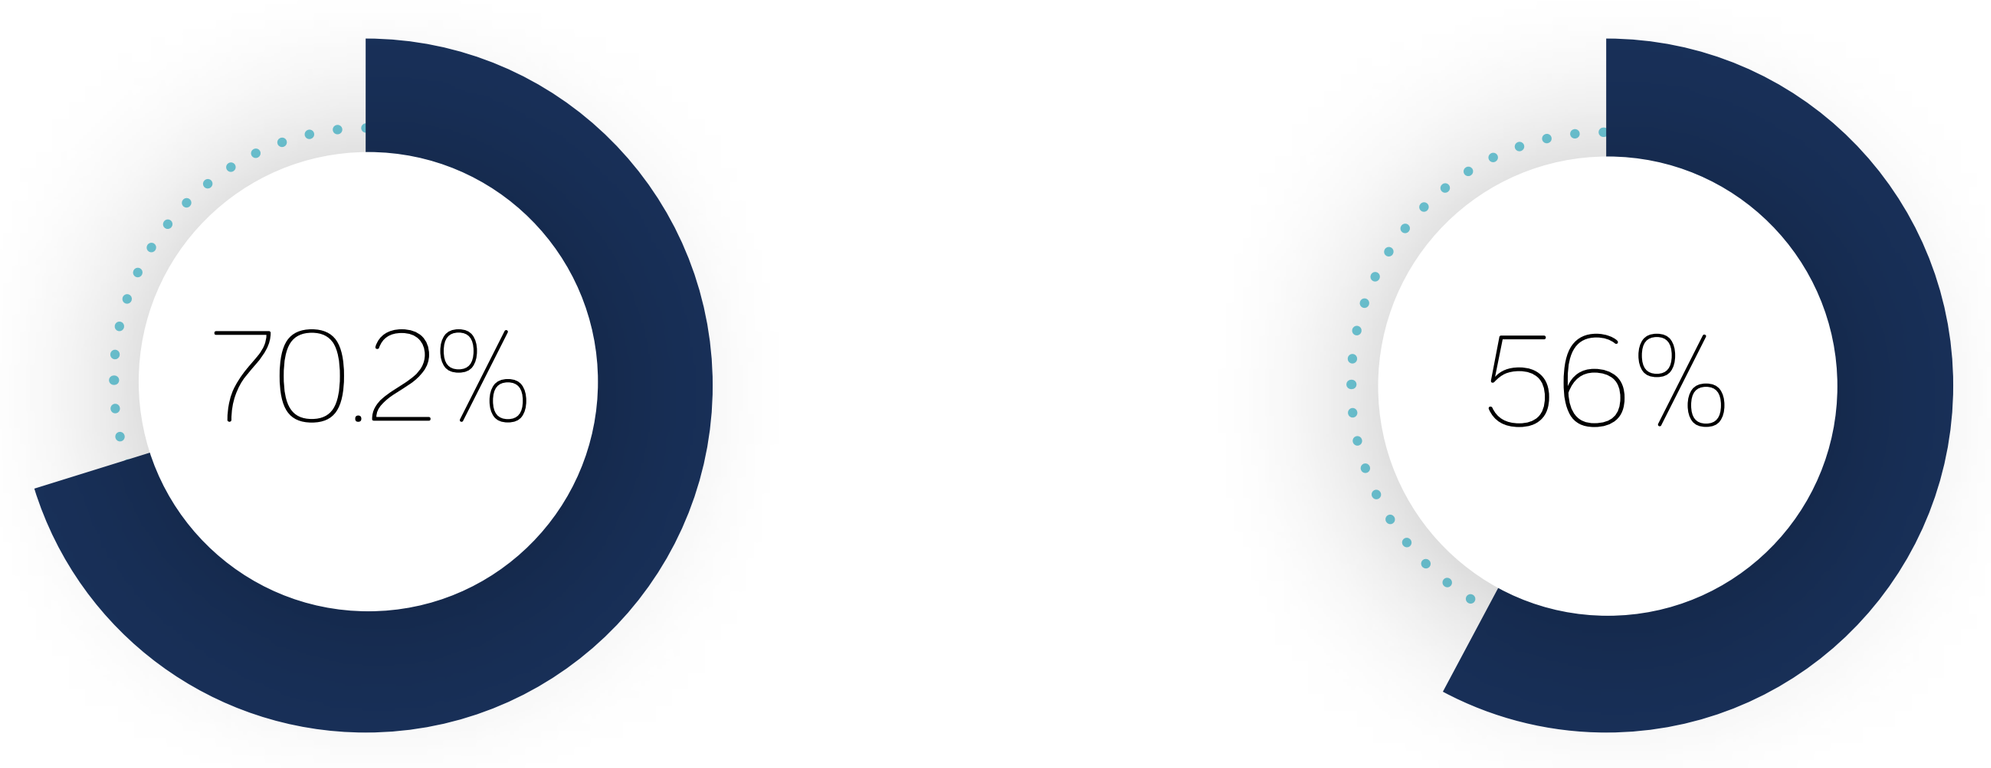
70.2%
56%

Average agreement rate between two pathologists on HER2 score

Kappa = 0.74 [0.72;0.76]

Rate of full consensus among all three pathologists on the HER2 score

Full consensus on HER2 score was reached for 253 cases out of 452

Figure 2 – Rate of pathologists' agreement in manual HER2 scoring. The agreement was calculated on the four HER2 scores of 0, 1+, 2+, 3+[1,2]

HER2 AI performance

The AI demonstrated high accuracy of HER2 scoring on the 419 validation slides

- The AI agreed with the majority vote in 82.1% (344) of the slides
- The AI agreed with at least one pathologist in 90.5% (379) of the slides
- The AI reached similar levels of accuracy across all labs and scanners

HER2 AI Score

Ground Truth Score	0	1+	2+	3+
0	67	16	0	0
1+	4	157	1	0
2+	0	48	36	5
3+	0	0	1	84

Figure 3 – Performance of the fully-automated HER2 algorithm vs. GT scores

When using different cutoffs for binary classification the resulting performance is:

HER2 Score classification	Kappa [95% CI]
0 vs. 1+/2+/3+	0.84 [0.76; 0.91]
0/1+ vs. 2+/3+	0.75 [0.68; 0.81]
0/1+/2+ vs. 3+	0.96 [0.92; 0.99]

Table 2 – The AI performance on different classification cutoffs

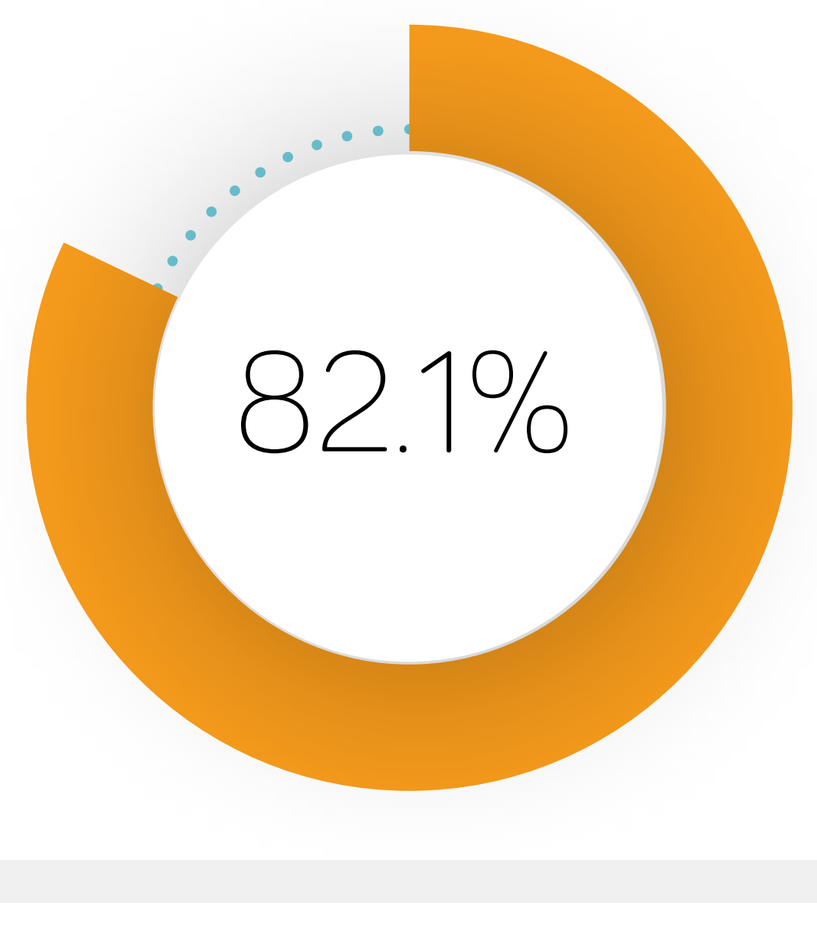
82.1%

Overall agreement between the AI HER2 score and the GT. (Kappa = 0.74, 95% CI [0.69; 0.80]). Quadratic Kappa between the AI score and the GT was 0.91 [0.89; 0.93].

HER2 stained cells analysis

High correlation between the percent of HER2 stained tumor cells, as detected by the AI, and the HER2 score distribution among the GT pathologists.

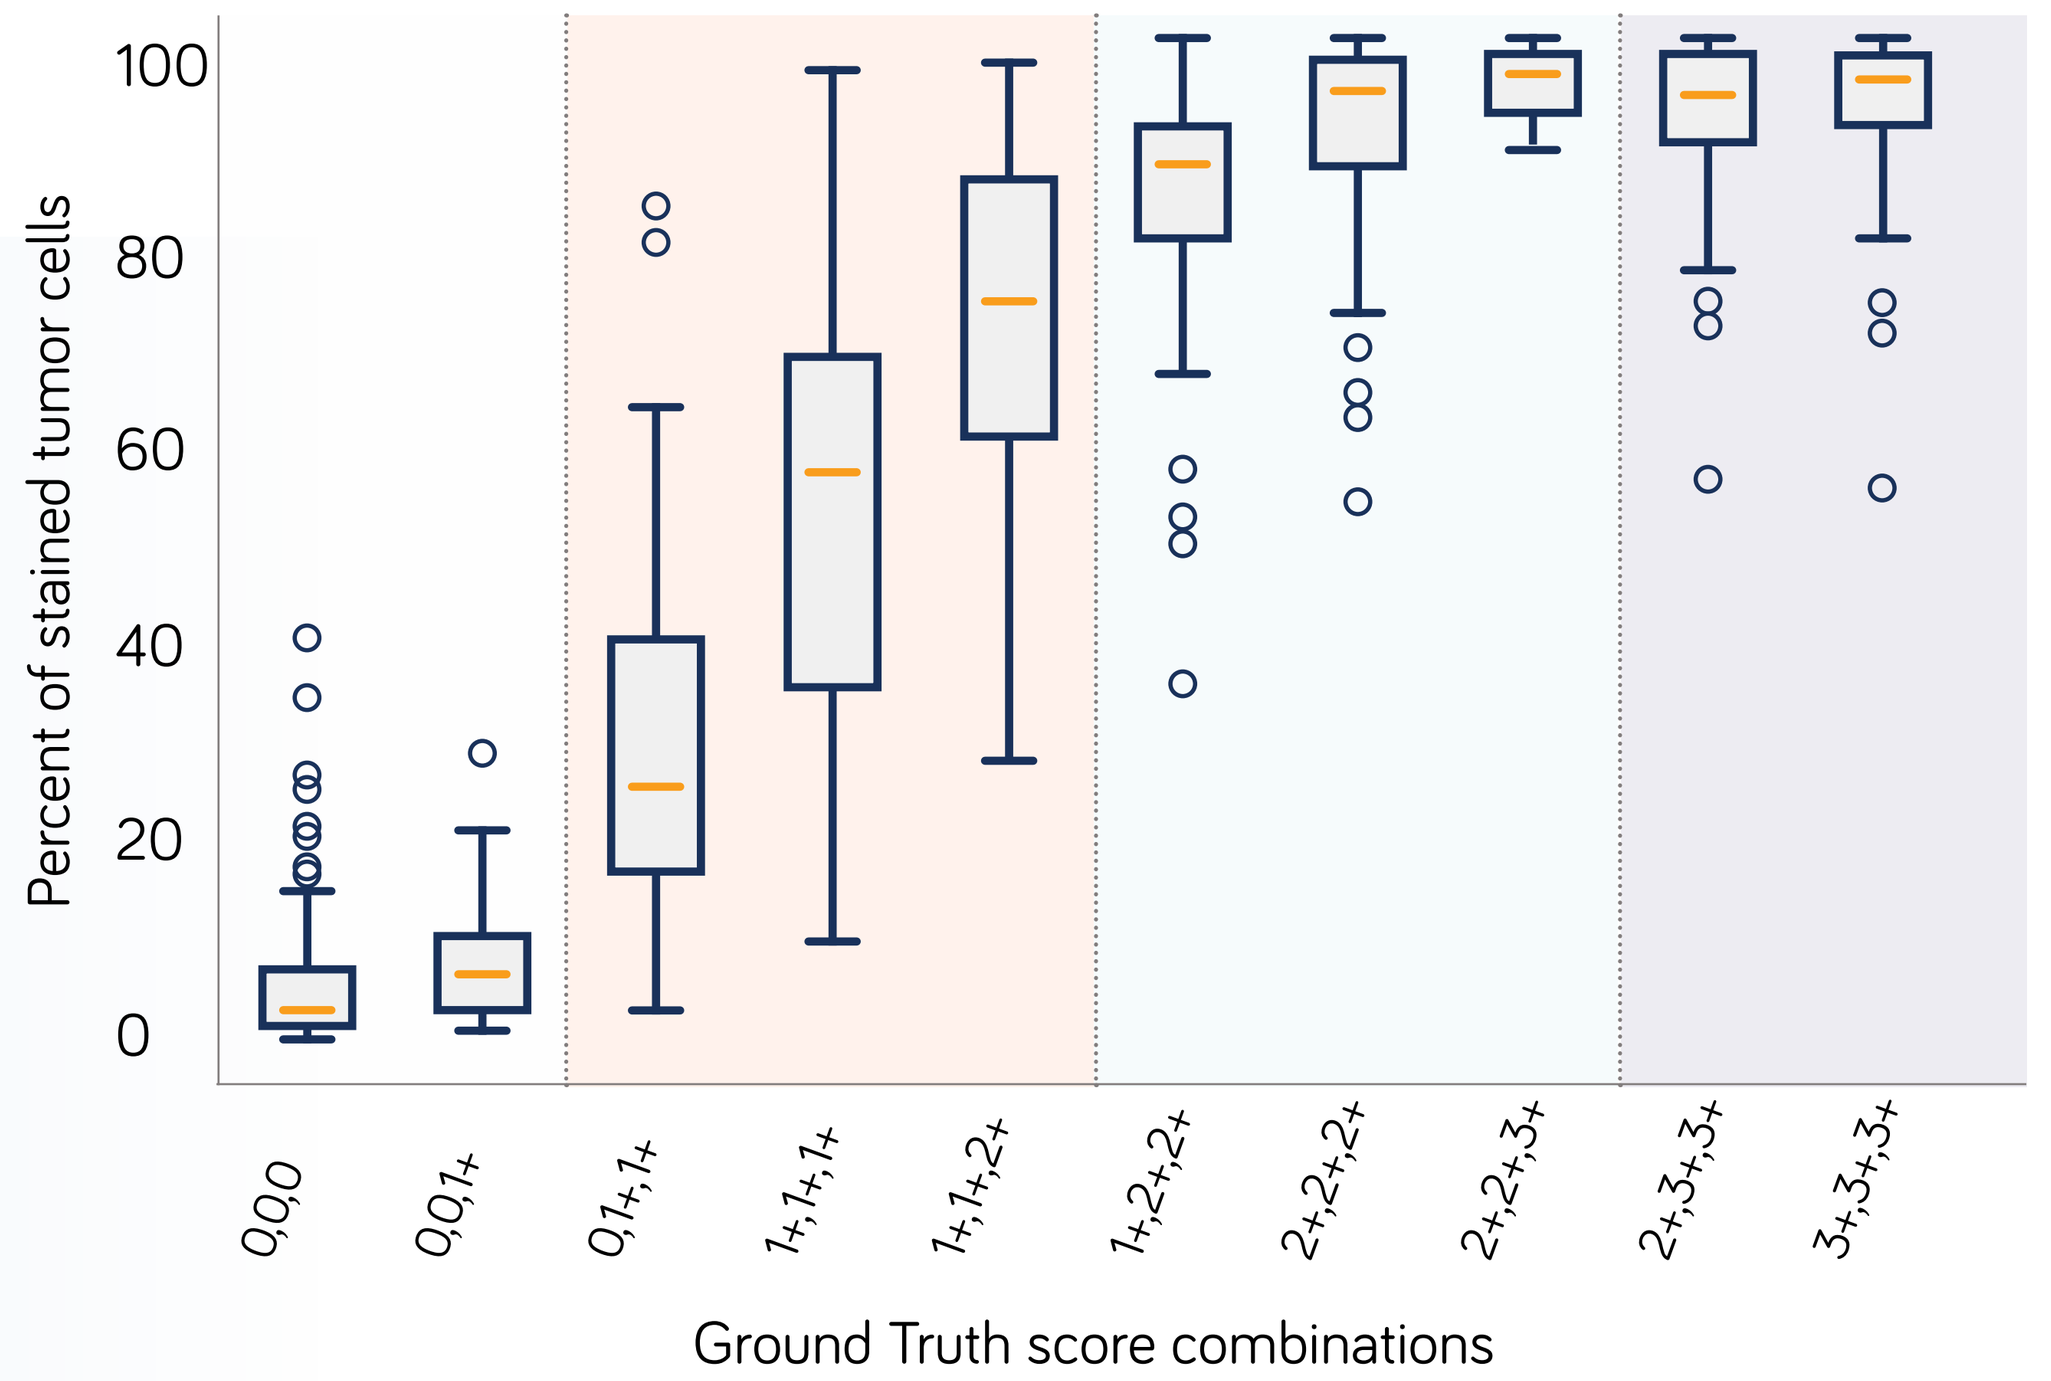
Percent of stained tumor cells
100
80
60
40
20
0
0,0,0
0,0,1+
0,1+,1+
1+,1+,1+
1+,1+,2+
1+,2+,2+
2+,2+,2+
2+,2+,3+
2+,3+,3+
3+,3+,3+
Ground Truth score combinations

Figure 4 - Box plot of the percent of stained tumor cells detected by the AI for each combination of GT scores

Examples of discrepancies between AI and GT

In many of the discrepancies between the AI and the GT, the AI gave a score that was given also by one of the GT pathologists. Here are two such examples:

A | GT HER2 = 0,0,1+, AI HER2 = 1+ **B |** Zoom-in on tumor cells

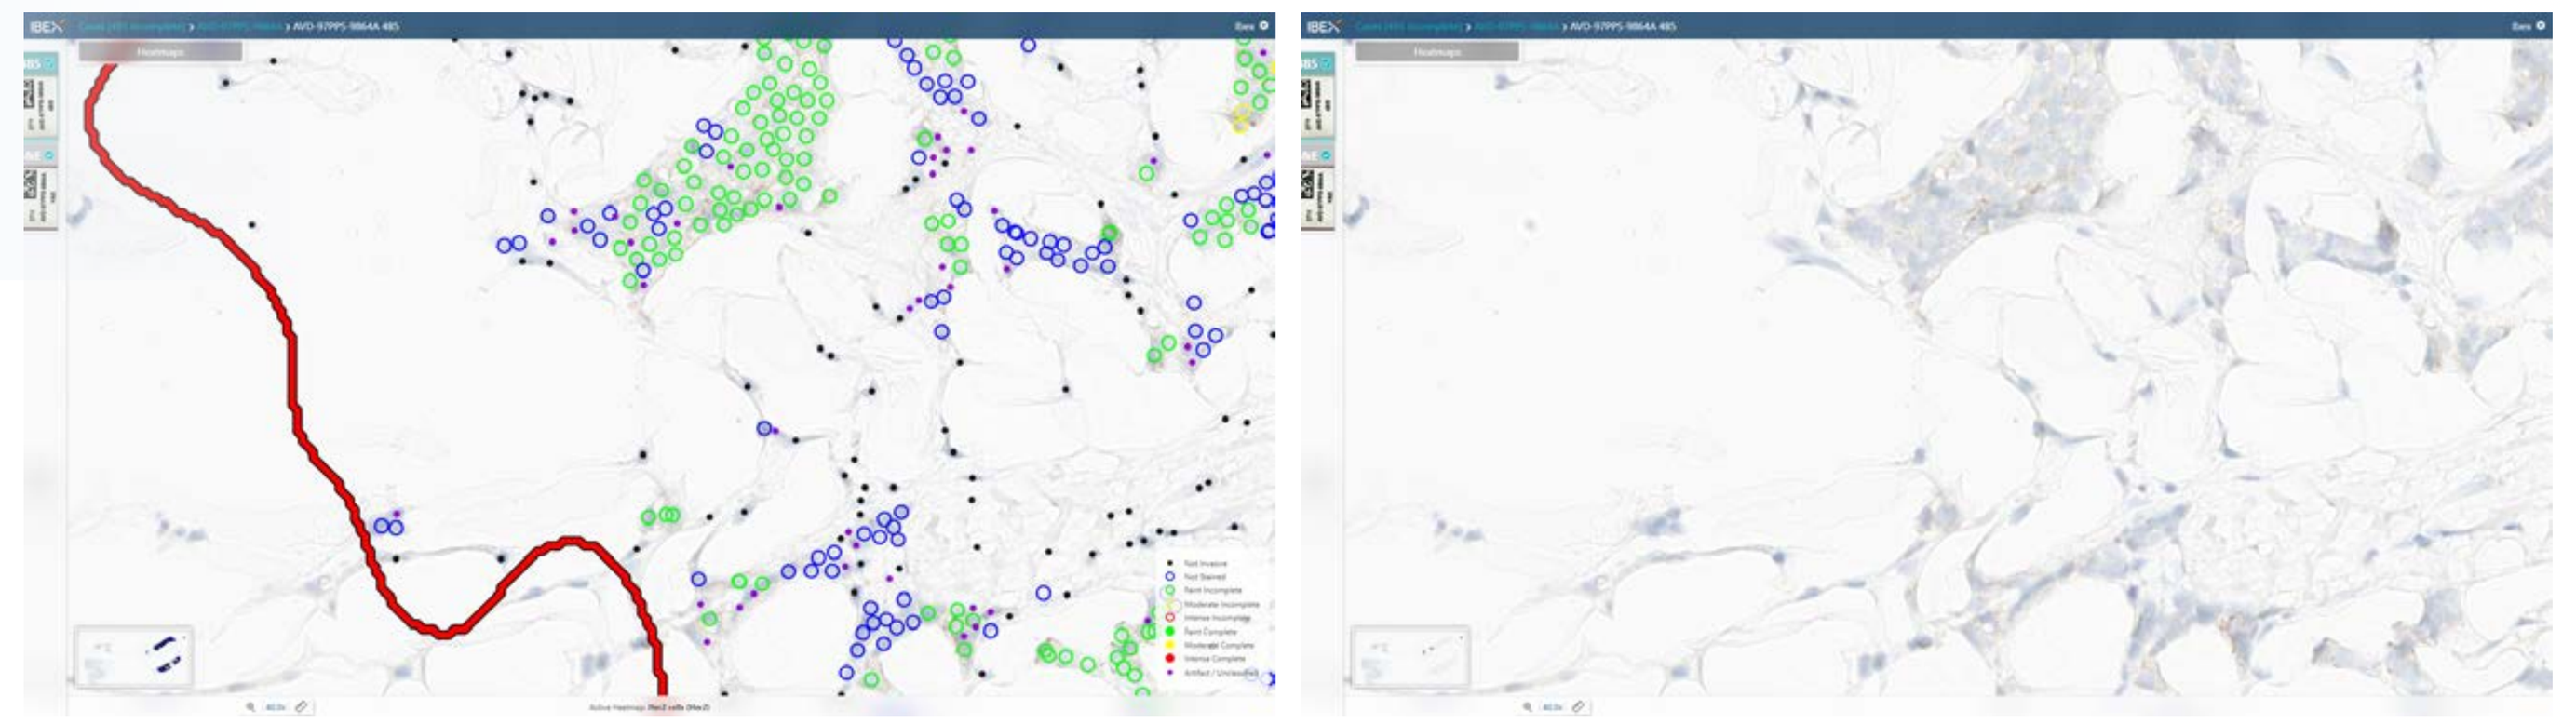

Figure 5 – The AI detects 19.7% stained cells, mostly faint incomplete, leading to a score of 1+. ROI at 40x magnification with AI cell classifications (A); ROI at 40x (B).

A | GT HER2 = 1+,2+,2+, AI HER2 = 1+ **B |** Zoom-in on tumor cells

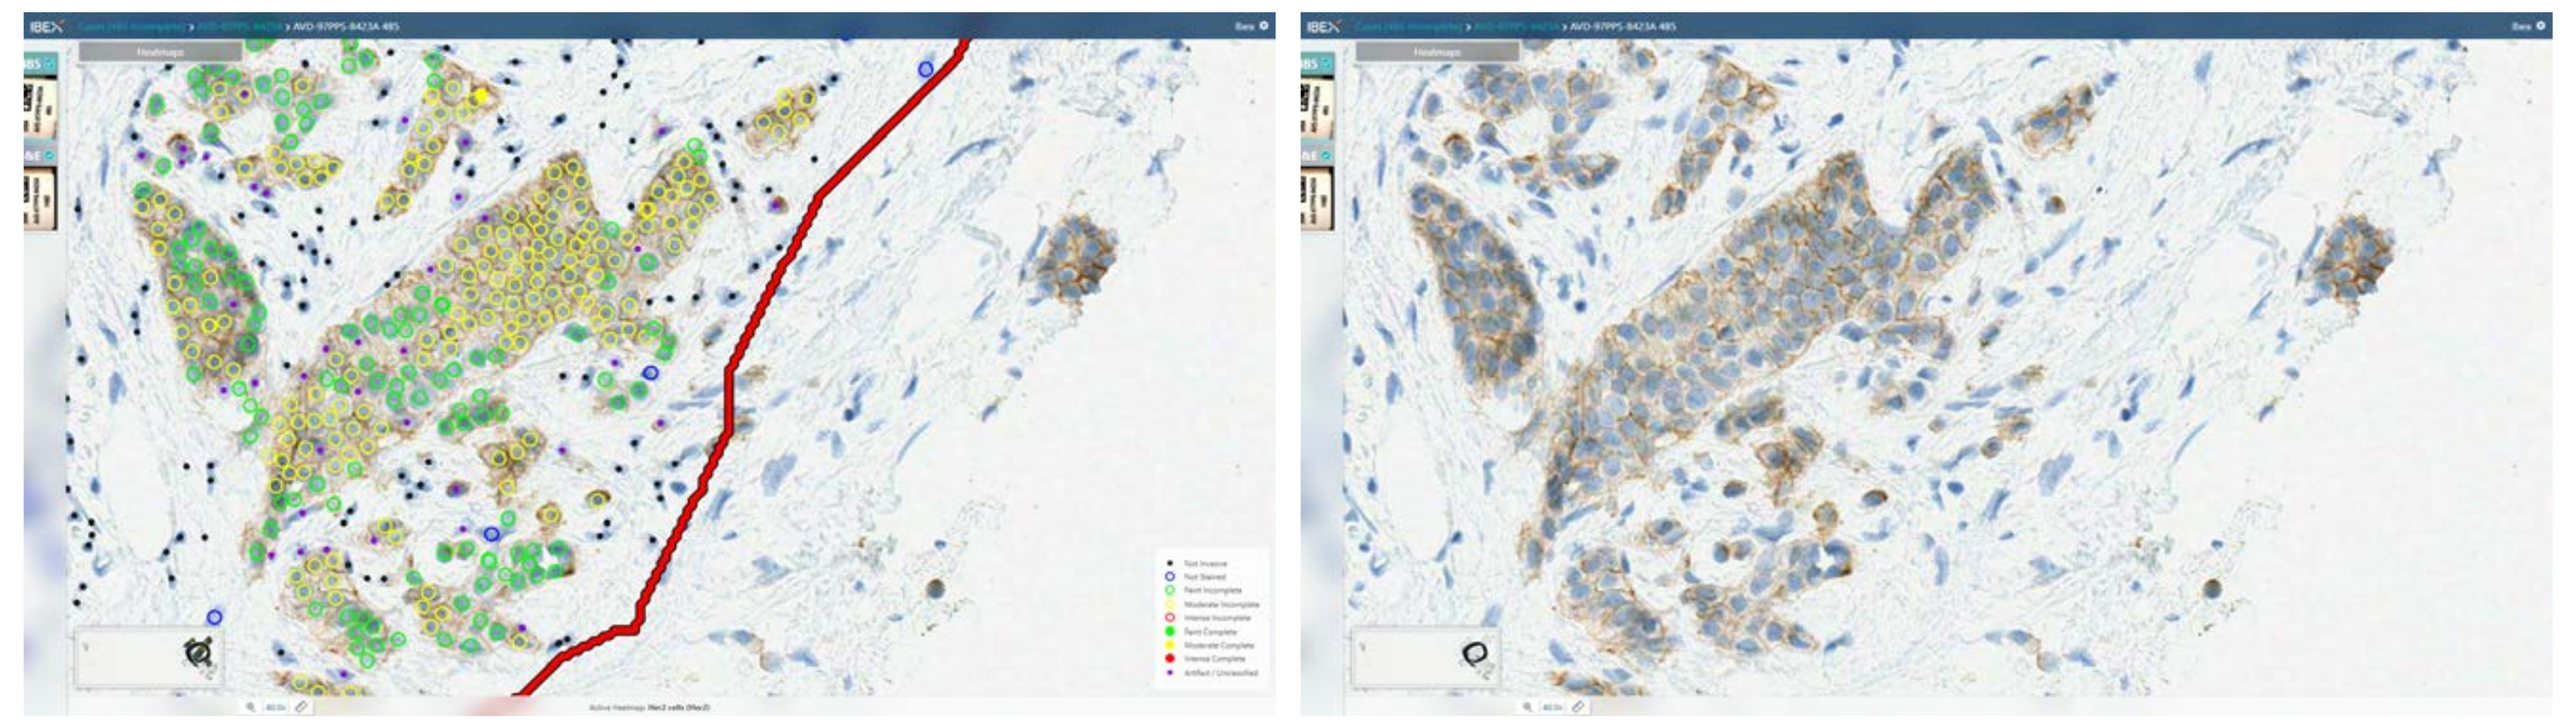

Figure 6 – The AI detects 13.5% not stained cells, 38.6% faint incomplete, 46.9% moderate incomplete, 1% moderate complete, leading to a score of 1+. ROI at 40x magnification with AI cell classifications (A); ROI at 40x (B).

CONCLUSIONS

- A fully automated AI-based solution for HER2 scoring was developed and tested.
- The study demonstrated that the AI solution automatically scores HER2 accurately, the findings being visualized to support pathologists' review.
- The HER2 AI solution demonstrated high accuracy (82.1%), especially in the context of a diverse and challenging validation dataset and rigorous review by 3 pathologists strictly according to the guidelines.
- AI solutions could be used as decision-support tools for pathologists in routine clinical practice, enhancing the reproducibility and consistency of HER2 scoring, thus enabling optimal treatment pathways and better patient outcomes.